IMAGE 1 (Immune Modulation and Gemcitabine Evaluation), a Randomised, Open-label Phase II Trial Comparing Gemcitabine with and without IMM-101 in Advanced Pancreatic Cancer

(Including results from the long-term follow-up Sub-Study)

Angus G Dalgleish† and the IMAGE 1 Trial Investigators‡ † St Georges, University of London ‡ See Acknowledgements

Survival in advanced pancreatic cancer

- The introduction of *nab*-paclitaxel/gemcitabine and FOLFIRINOX has improved median survival for patients with metastatic disease and good performance status (PS, ECOG 0-1) (8.7 months[1] and 11.1 months[2] respectively) compared to gemcitabine (GEM) monotherapy (typically 5.7-6.8 months)[2,3,4]
- Patients with lower PS (ECOG 2) and/or a comorbidity profile precluding more aggressive regimes have fewer options with GEM monotherapy recommended, both as first and second line treatment[5]
- Long-term survival remains elusive - 4% at 36 months for patients treated with *nab*-paclitaxel/GEM in the phase 3 MPACT study[1] with only 1% patients in the 'at risk' category at 36 months in the FOLFIRINOX arm of the phase 3 PRODIGE 4/ACCORD trial[2]
- Five-year survival rates for subjects diagnosed with distant disease remain low at 3%[6]
- Toxicity to treatment limits exposure time: 22% of patients withdrew from the *nab*-paclitaxel/GEM arm of the MPACT trial due to unacceptable treatment-related toxicity[1] and neutropenia ≥ grade 3 occurred in 38% of patients treated with nab-paclitaxel/GEM[1] and 46% treated with FOLFIRINOX[2]

The IMAGE 1 Trial

- Open-label phase 2 study (NCT01303172) comparing safety and efficacy of GEM with and without IMM-101 in subjects with advanced (unresectable stage 3 or stage 4) pancreatic cancer[7]
- Patients randomised 2:1 to IMM-101 (0.1mL intradermal injection of 10mg/mL suspension) + GEM (1000mg/m^2) or GEM
- Study treatment could be continued to a maximum of 12 cycles of GEM in the Main Study
- Patients from both treatment groups who completed the Main Study were eligible to take part in a long-term follow-up study (Sub-Study) where all received IMM-101 and, at the Investigator's discretion, adjunctive chemotherapy
- Efficacy was assessed with Kaplan-Meier curves and log-rank test
- All statistical testing was at 2-sided 5% significance level with hazard ratios and p-values from Cox PH regression models

Patients

	ITT		ITT Metastatic Subgroup	
	IMM-101 + GEM	GEM	IMM-101 + GEM	GEM
n	75	35	64	28
Age, median (range)	68 (45-88)	66 (53-83)	68 (45-88)	65.5 (53-83)
% male	51	60	NC	NC
ECOG 0-1	62 (83%)	32 (91%)	55 (86%)	26 (93%)
ECOG 2	13 (17%)	3 (9%)	9 (14%)	2 (7%)
Completed Main Study	12 (16%)	1 (3%)	12 (19%)	0
Months on Study*, median (range)	4.83 (0.16-46.49)	2.79 (0.46-12.75)	NC	NC

NC = not calculated *Main Study and Sub-Study combined

The Investigational Product: IMM-101

- Suspension of heat-killed whole cell *Mycobacterium obuense* (NCTC13365) in borate-buffered saline
- Administered by intradermal injection to the upper arm, alternate arms for each dose
- Myeloid dendritic cell (DC) activator, enhancing antigen processing
- Produces Type-1 immune response, with formation and activation of CD8$^+$ Cytotoxic T-Lymphocytes (CTLs) and increased production of the cytokine interferon-γ (IFN-γ)
- Matures monocytes into Type-1 macrophages (M1) and increases the number of activated IFN-γ producing Th1, NK, γδ-T and NKT cells
- CTLs, M1, NK, γδ-T cells and NKT cells and IFN-γ are known to play crucial roles in anti-tumour responses, attacking the tumour through different but complementary pathways

ITT (All Patients)

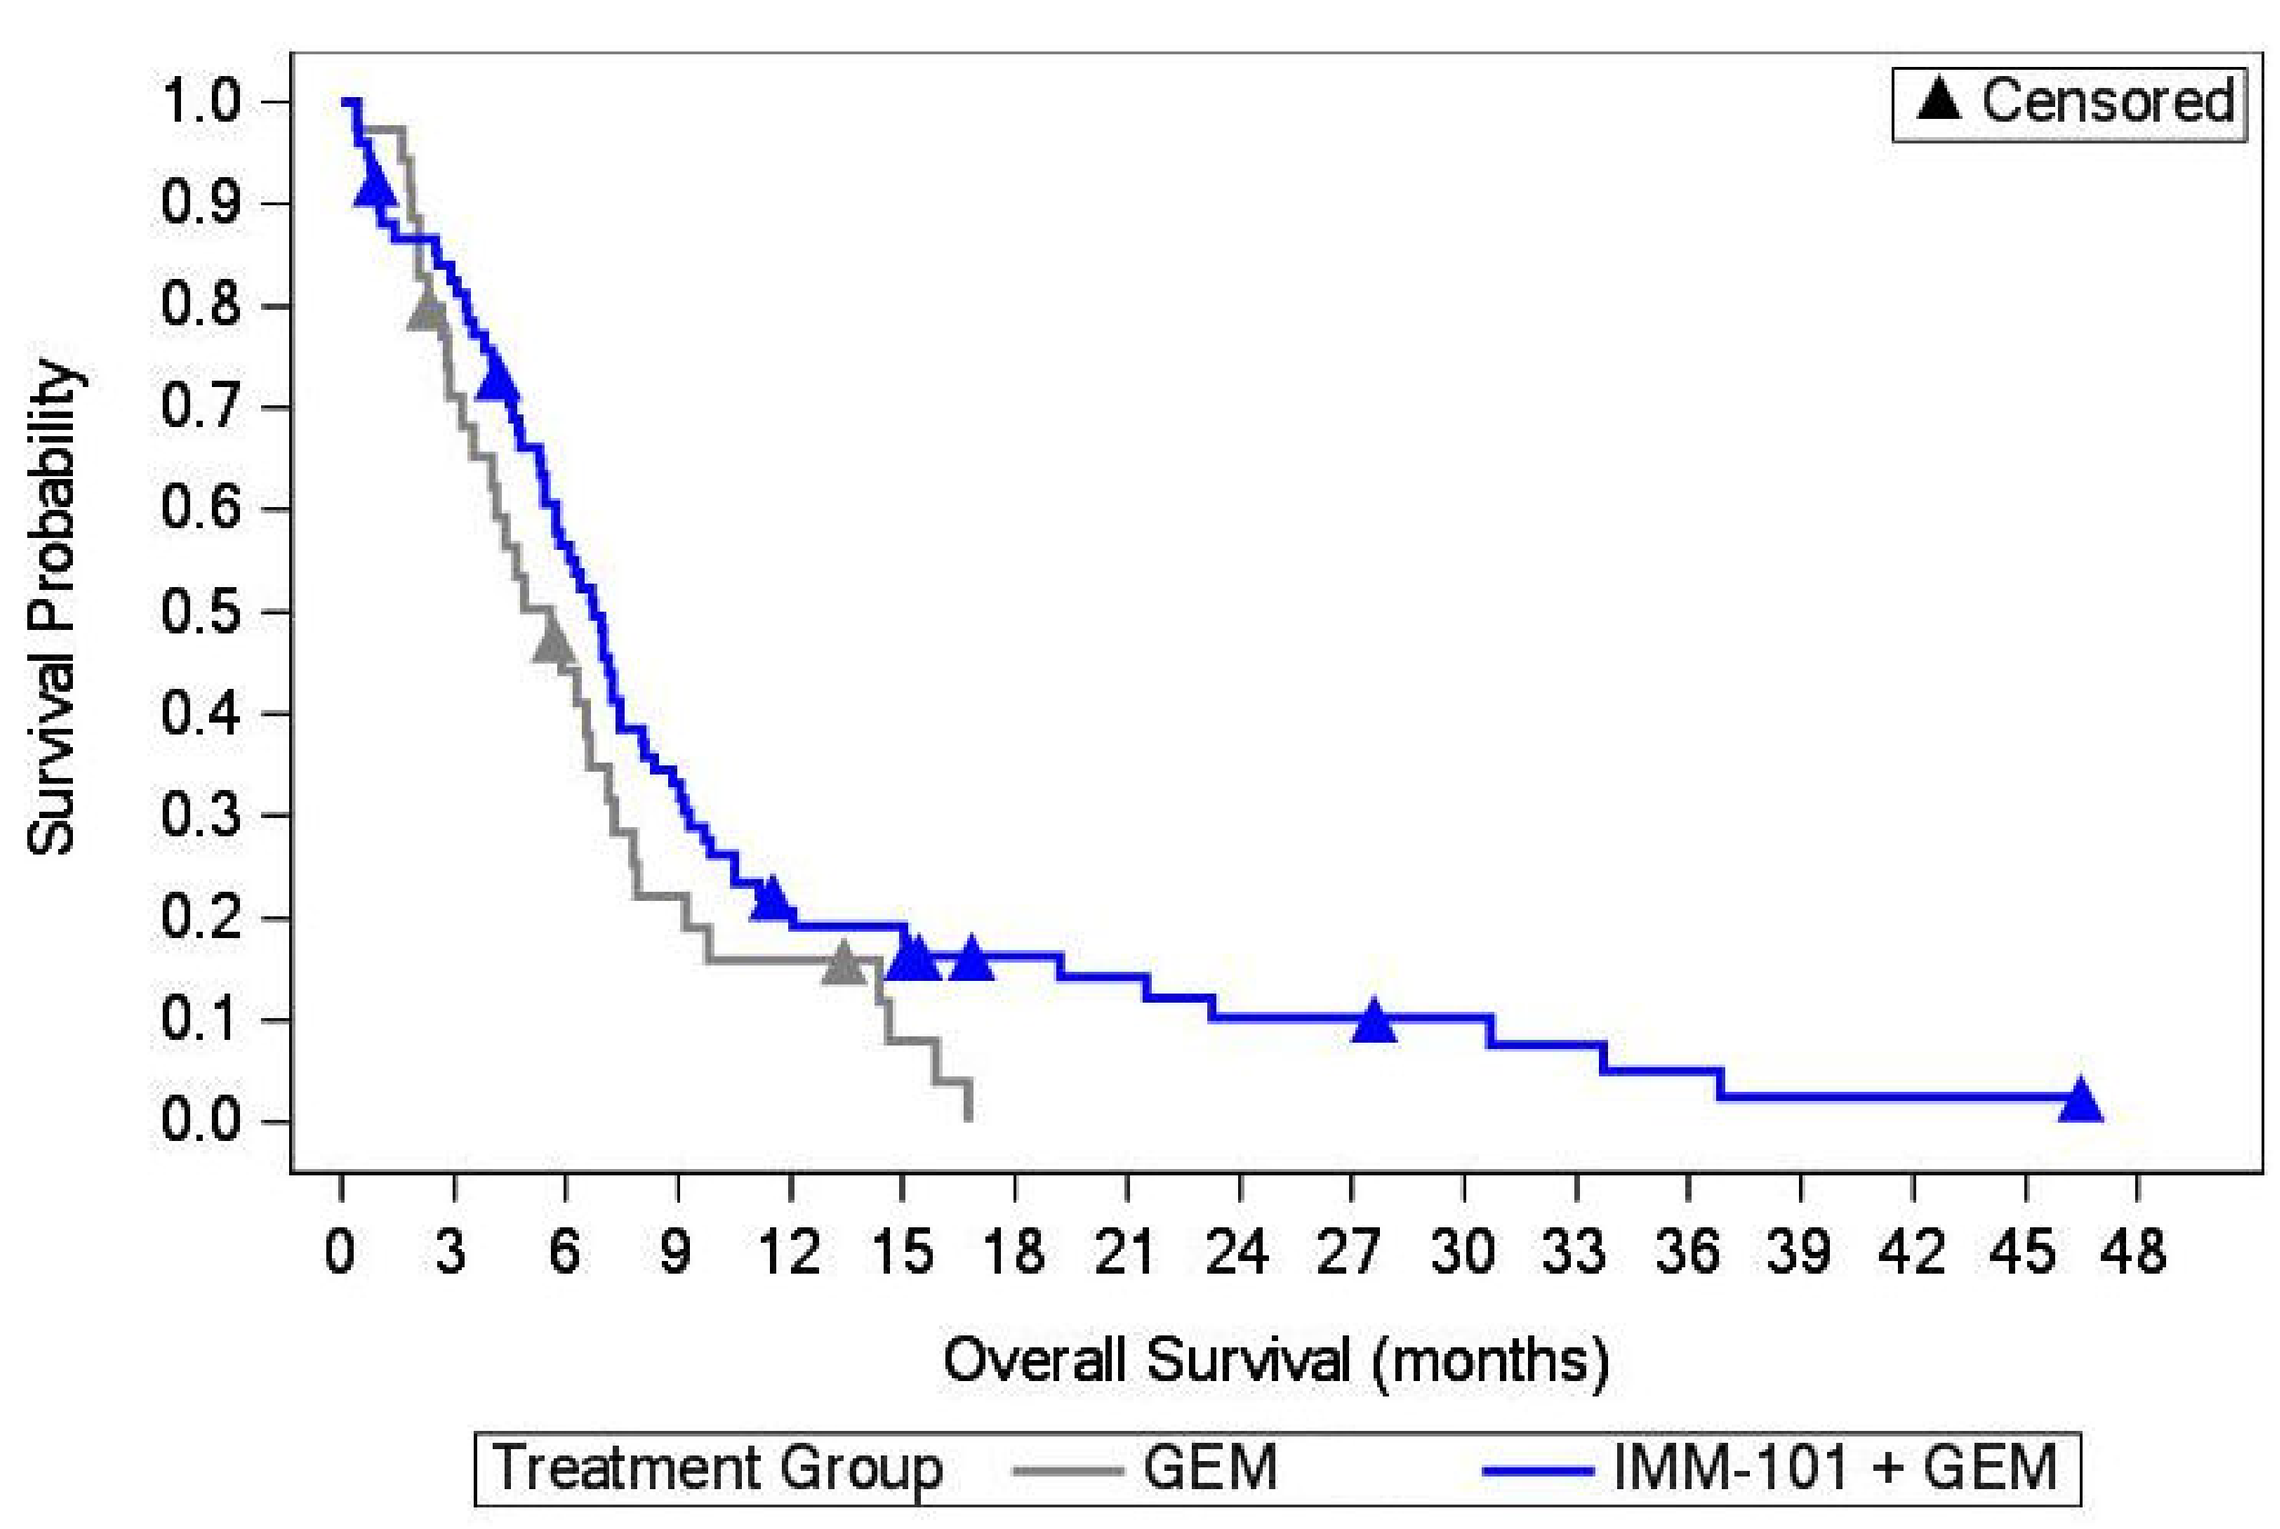
Censored
Survival Probability
Overall Survival (months)
Treatment Group GEM IMM-101 + GEM

Overall Survival Results

ITT			ITT Metastatic Subgroup	
IMM-101 + GEM n = 75	GEM n = 35		IMM-101 + GEM n = 64	GEM n = 28
67 (89.3)	32 (91.4)	Number of events n (%)	56 (87.5)	26 (92.9)
8 (10.7)	3 (8.6)	Number censored n (%)	8 (12.5)	2 (7.1)
6.7 (5.4-7.5)	5.6 (3.2-7.2)	Median OS: months (95% CI)	7.0 (5.5-9.0)	4.4 (2.8-6.5)
0.0706		Log-rank p-value	0.0093	
0.67 (0.44-1.04)		HR (95% CI)	0.53 (0.33-0.86)	
		Probability of survival: % (95% CI)		
20.3 (12.0-30.3)	**15.8** (5.8-30.1)	12 month	**24.0** (14.2-35.2)	**11.5** (2.9-26.5)
15.4 (8.1-24.9)	**1.8** (0.0-12.8)	18 month	**18.3** (9.6-29.1)	**2.3** (0.0-15.7)
9.7 (3.9-18.6)	NA	24 month	**11.4** (4.6-21.7)	NA
9.7 (3.9-18.6)	NA	30 month	**11.4** (4.6-21.7)	NA
4.8 (1.0-13.4)	NA	36 month	**5.7** (1.2-15.6)	NA

NA = not available as no survival data at this point

Metastatic Sub-Group

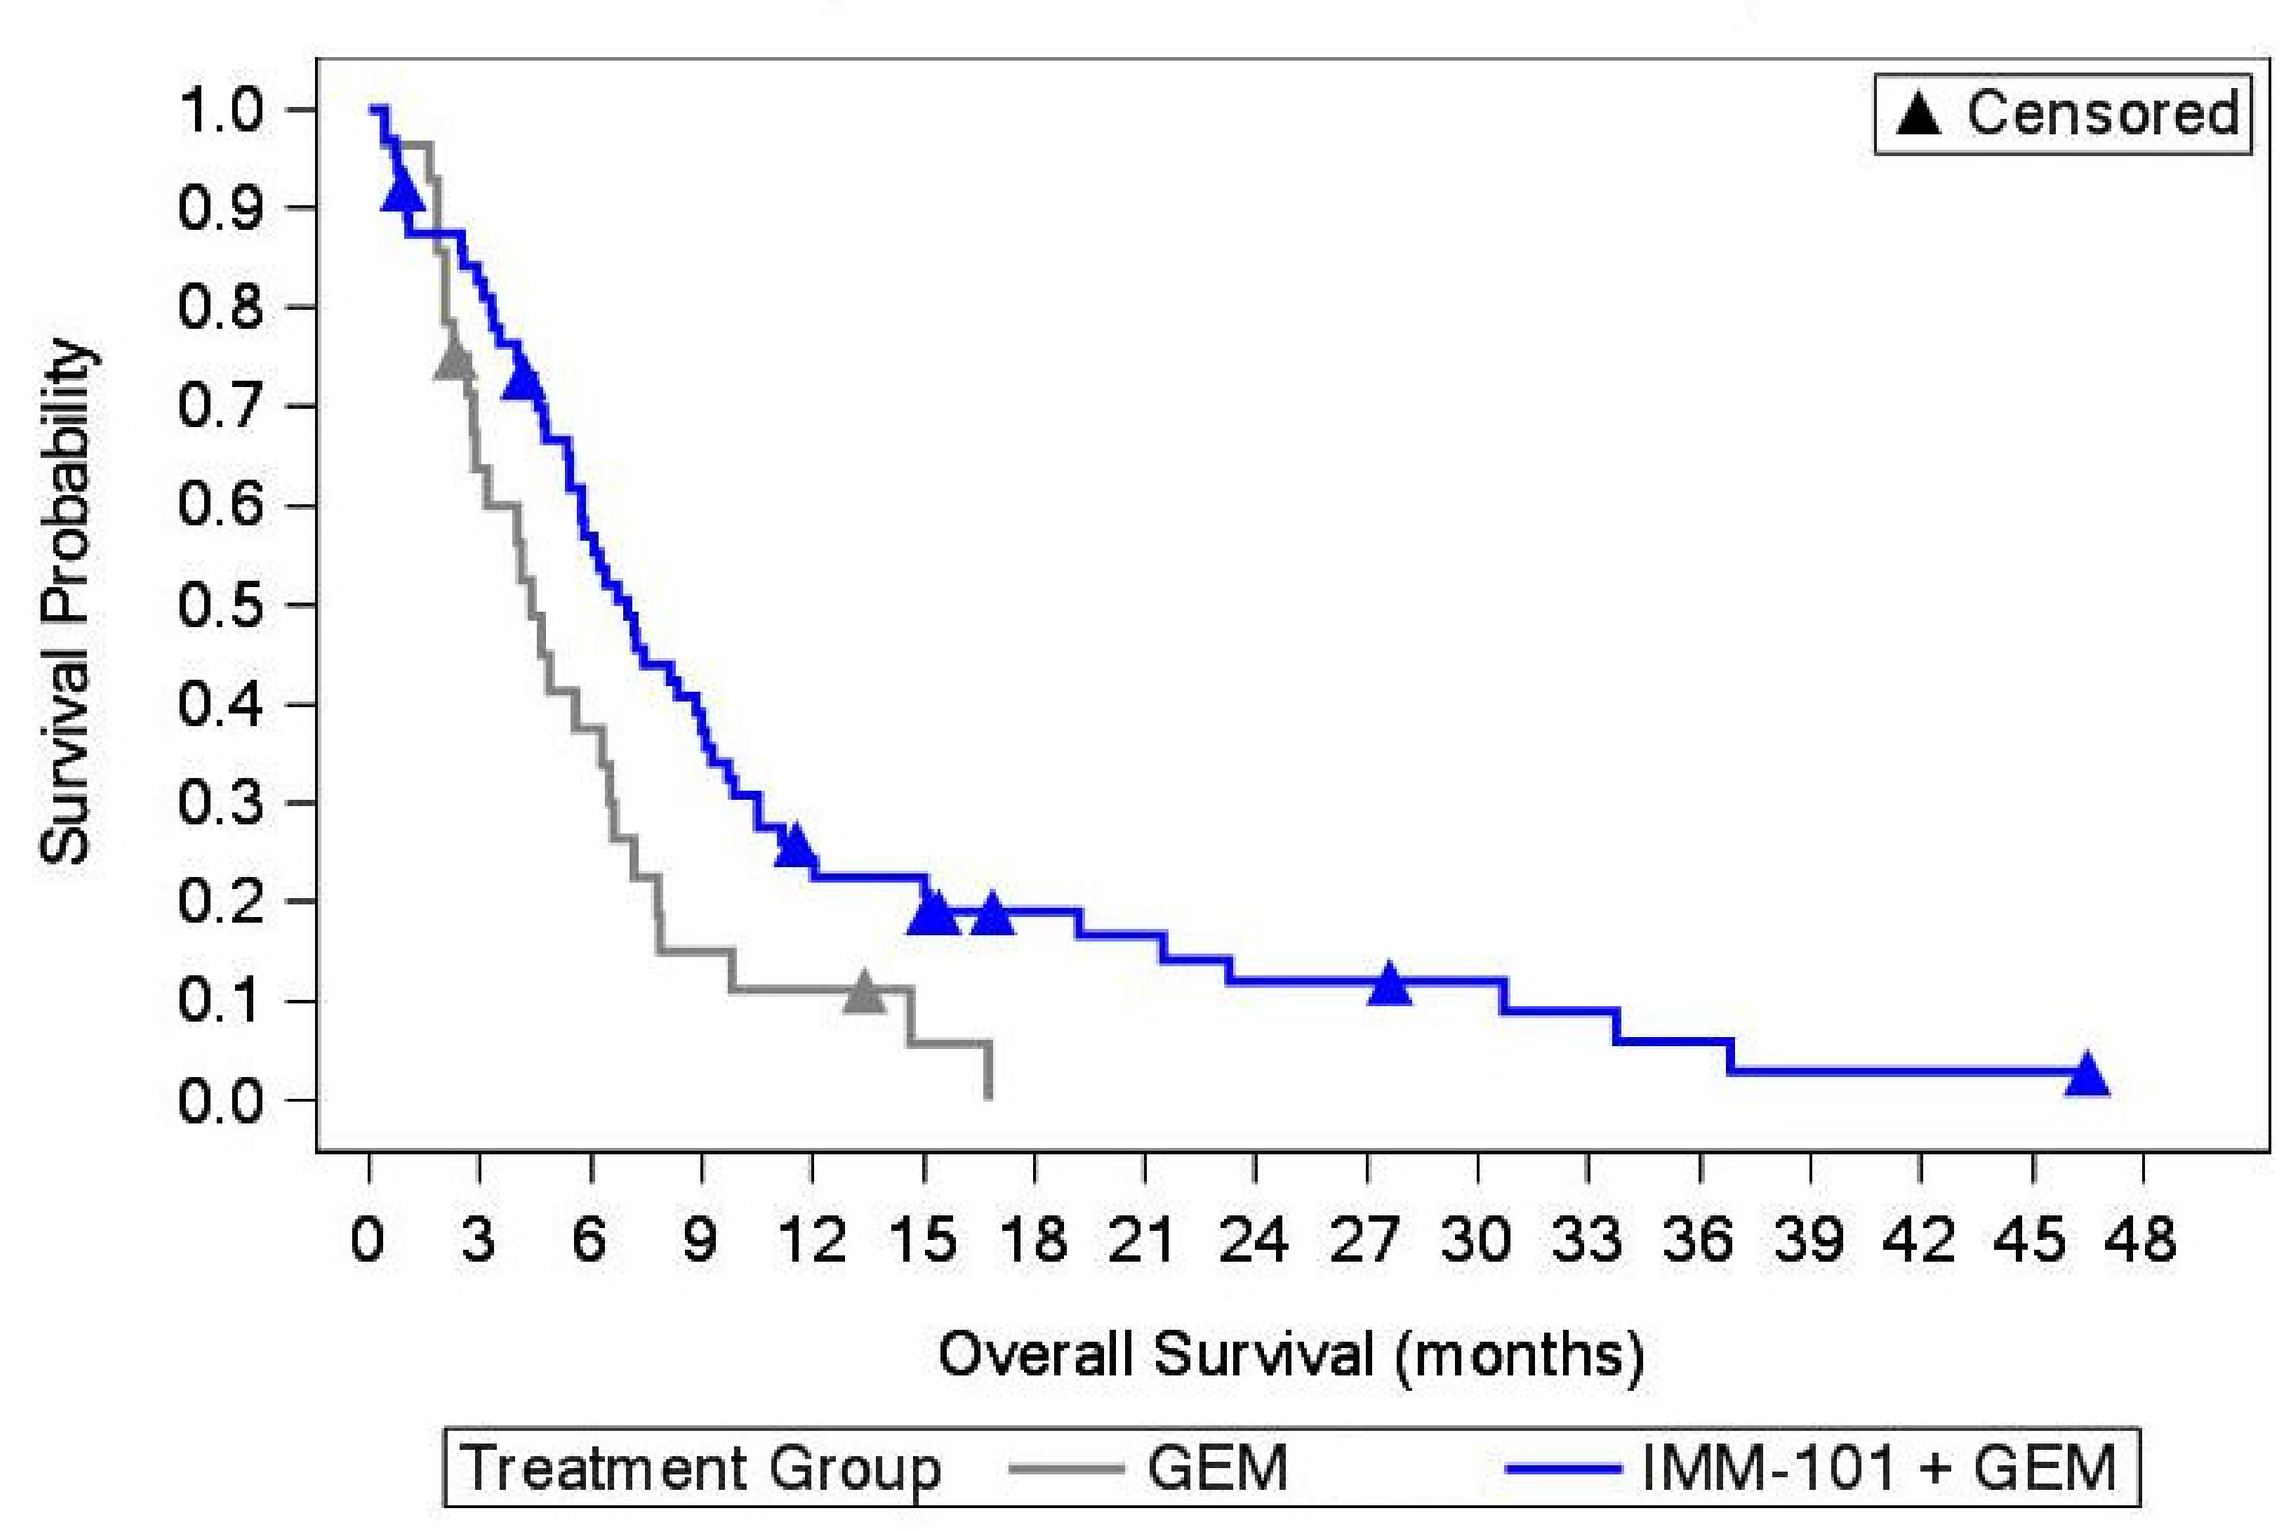
Censored
Survival Probability
Overall Survival (months)
Treatment Group GEM IMM-101 + GEM

Frequently Asked Questions

- The results indicate a benefit for the IMM-101 + GEM group but the median OS values are low compared to other studies – why?

 Median OS for the ITT GEM group is similar to results from other studies for patients randomised to GEM (e.g. erlotinib trial[4]). OS in trials of metastatic patients can vary depending on demographics and was only 4.3 months for patients with Karnofsky 70-80 (ECOG 1) in the MPACT trial[1]. Phase 3 trials usually limit recruitment to newly diagnosed patients, whereas IMAGE 1 had no limit to the time from diagnosis (All ITT patients 0.1-6.9 months for IMM-101 + GEM group; 0.1-3.9 for GEM).
- Do you think any imbalance in baseline patient characteristics could account for the survival differences?

 Exploratory univariate analyses (Cox PH regression model) showed the HR to be maintained when factors of CA19.9, CEA, LDH, CRP, NLR, total bilirubin, age and PS were accounted for, indicating baseline factors were not responsible. Exploratory multivariate stepwise Cox PH regression analyses supported this conclusion[7,8].
- How do the long-term survival probability figures compare with those from other studies?

 Long-term survival from the MPACT study showed 10% survival probability at 24 months and 4% at 36 months for the nab-paclitaxel/GEM group, with 5% at 24 months and 0% at 36 months for the GEM group[1]. IMAGE 1 results compare favourably although it should be borne in mind that IMAGE 1 was a proof of concept study and genuine treatment differences were not planned to be detected with high power.
- Which chemotherapies did patients in the Sub-Study receive?

 Eight of the 12 patients took chemotherapies with capecitabine the most frequently used (5 patients). Others were GEM/paclitaxel (3 patients), GEM and paclitaxel alone (2 patients) and combinations or sole use of oxaliplatin, 5-fluorouracil and irinotecan (3 patients). The number of different chemotherapies taken ranged from 1 to 4.
- How could these have influenced your results?

 Long-term survival results are usually influenced by subsequent therapy, taken off study, with 38-50% second-line therapy used in treatment arms of recent phase 3 trials[2,9]. However, for IMAGE 1, patients continued on study so they could still receive IMM-101.

Safety and Tolerability

Overall Exposure to IMM-101 (months)			
	IMM-101 + GEM		GEM
	Combined Studies	Sub-Study	Sub-Study
n*	74	11	1
Mean (StD)	6.24 (7.520)	8.56 (9.785)	1.22 (NE)
Median	3.73	4.60	1.22
Range	0.03-46.49	0.95-34.30	1.22-1.22

* Number of patients receiving IMM-101
NE = Non-evaluable

- Safety data for the Main Study is already published[7]. Completion of the Sub-Study has allowed the safety profile of IMM-101 to be further assessed during long-term exposure patients entering the Sub-Study.
- No patients withdrew from the Sub-Study due to adverse events.
- Maximum exposure to IMM-101 was 34.3 months in the Sub-Study and 46·5 months over the combined Main and Sub studies.
- 8 patients experienced 31 AEs that were grade ≥ 3 in the Sub-Study. None of these events was experienced by more than 1 patient. None were related to IMM-101 except a procedure of pancreaticduodenectomy on one patient, performed due to regression of the primary tumour, considered a benefit of treatment.
- 3 patients experienced 7 SAEs during the Sub-Study. None were considered related to IMM-101.
- 4 patients experienced 11 AEs that were considered related to IMM-101 during the Sub-Study. The majority of these were injection site reactions (see Table).
- 8 of the 12 Sub-Study patients received only full doses of IMM-101 during the combined Main and Sub-Study periods. The other 4 patients received some half doses due to local reactions.

IMM-101 Related AEs in Sub-Study	
Preferred term (MedDRA)	IMM-101 + GEM (n=11)[1] n (E)
Injection site reaction	2 (5)
Injection site erythema	1 (1)
Injection site induration	1 (1)
Injection site laceration	1 (1)
Axillary mass	1 (1)
Diarrhoea	1 (1)
Pancreaticduodenectomy	1 (1)

[1] No AEs reported as related to IMM-101 for the patient from the GEM group of the Main Study
n= number of patients, E= number of events

Acknowledgements

CHIEF INVESTIGATOR	DALGLEISH A G,	St George's University of London, UK
SPAIN	DIAZ R	Hospital La Fe de Valencia, Valencia,
	FERNANDEZ-MARTOS C	Instituto Valenciano de Oncologia, Valencia
	MASSUTI B	Hospital General de Alicante, Alicante
	MUÑOZ A	Hospital Gregorio Marañon, Madrid
	PAZO R	Hospital Miguel Servet, Zaragoza
	VIEITEZ DE PRADO J-M	Hospital Central de Asturias, Asturias
UK	ADAMSON D	Ninewells Hospital, Dundee
	ARIF S	Velindre Cancer Centre, Cardiff
	CHANG D and MEHTA A,	Blackburn Royal Infirmary, Blackburn
	CHEESEMAN S	Bradford Royal Infirmary, Bradford, and Airedale General Hospital, Keighley
	GLYNNE-JONES R	Mount Vernon Cancer Centre, Northwood
	McADAM K	Peterborough City Hospital, Peterborough
	STEBBING J	The London Clinic Cancer Centre, London
ITALY	ALABISO O	Azienda Ospedaliero-Universitaria Maggiore della Carità, Novara
	BIDOLI P	Azienda Ospedaliera San Gerardo, Monza
	Di MARCO M	Azienda Ospedaliero-Universitaria Orsola Malpighi, Bologna
	GRANETTO C	Azienda Ospedaliera Santa Croce e Carle, Cuneo
	ZANIBONI A	Fondazione Poliambulanza, Brescia
CYPRUS	PAPAMICHAEL D,	Bank of Cyprus Oncology Centre, Nicosia
IRELAND	McDERMOTT R,	St Vincent's University Hospital, and The Adelaide and Meath Hospital, Dublin.

We thank the patients who volunteered to participate in this study, the Investigators and staff members at the study sites, the members of the Data Monitoring Committee and representatives of the Sponsor who were involved in data collection and analyses. We also thank Exploristics Limited for data management and statistical support.

The IMAGE 1 study was supported by Immodulon Therapeutics and this poster and related information can be found at www.immodulon.com

Conclusions

Conclusions from the completed IMAGE 1 Sub-Study:

- The IMAGE 1 Sub-Study identified long-term survivors from the Main Study and showed IMM-101 to be well tolerated over an extended period, with a maximum exposure of 46.5 months.
- The survival probability at 24, 30 and 36 months indicated long-term benefits of the previously reported treatment effect for IMM-101 used with GEM and, in some cases, with a variety of other anti-cancer treatments. The figures at 24 and 36 months were similar to those seen with nab-paclitaxel/GEM[1].
- No additional safety signals were identifiable from the Sub-Study.

Future Directions:

IMAGE 1 demonstrated promising efficacy and tolerability of IMM-101 + GEM with the possibility of long-term use and survival benefits. There is potential for use in metastatic pancreatic cancer as:

- First line treatment in patients who are unsuitable for more aggressive regimes
- Second-line/maintenance therapy

References

1. Goldstein D *et al*., J. Natl. Cancer Inst. 2015, 107 (2), dju413
2. Conroy T *et al*., N. Engl. J. Med., 2011, 364, 1817-25
3. Burris HA III *et al*., J. Clin. Oncol., 1997, 15, 2403-13
4. Moore MJ *et al*., J. Clin. Oncol., 2007, 25 (15), 1960-66
5. Sohal DPS *et al.,* J. Clin. Oncol., 2016, 36 (24), 2545-56
6. Cancer facts and figures, American Cancer Soc., 2018
7. Dalgleish AG *et al.,* Br. J. Cancer 2016, 115, 789-96
8. Immodulon Therapeutics IMM-101 Investigator's Brochure, 2018, Ed. 13
9. Von Hoff DD *et al.,* N. Engl. J. Med. 2013;369(18):1691-1703

Presented at the 33rd Annual Meeting of the Society for Immunotherapy of Cancer, Washington DC, USA, November 2018