LONG-TERM SURVIVAL IN IMAGE 1: A MULTI-CENTER RANDOMIZED, OPEN-LABEL, PROOF OF CONCEPT, PHASE 2 TRIAL COMPARING GEMCITABINE WITH AND WITHOUT IMM-101 IN ADVANCED PANCREATIC CANCER

ANGUS G. DALGLEISH AND THE IMAGE 1 TRIAL INVESTIGATORS, St George's, University of London, London, UK

BACKGROUND

- Immunotherapy may produce durable responses in some cancers.
- There is an urgent need for pancreatic cancer treatments that improve survival without adversely affecting safety and quality of life.
- IMM-101 is a systemic immunomodulator that induces protective CD8+ T cell responses and reduces metastatic burden in murine models of pancreatic cancer (Elia *et al.*, 2013).
- IMM-101 is a suspension of heat-killed whole cell *Mycobacterium obuense* (NCTC13365) in borate-buffered saline, administered intradermally.
- It has been reported that IMM-101 is associated with clinically meaningful increases in OS and PFS in patients with metastatic pancreatic cancer and does not appear to confer an incremental safety burden beyond that associated with chemotherapy and the disease itself (Dalgleish *et al.*, 2015).
- Here we report long-term survival in patients with advanced pancreatic cancer receiving IMM-101 enrolled in the IMAGE 1 clinical trial.

METHODS

- Proof of concept Phase 2 study (NCT01303172) to direct future development of IMM-101 in pancreatic cancer.
- Patients were randomly assigned in a 2:1 ratio to receive IMM-101 (0.1mL intradermal injection of 10mg/mL) + Gemcitabine (Gem, 1000mg/m^2) (IMM-101 treated group) or Gem alone (Control group).
- Per protocol this could be continued to a 12-cycle maximum.
- Efficacy was assessed with Kaplan-Meier curves and the log rank test. All statistical testing was at 2-sided 5% significance level. Hazard ratios and p values are from Cox regression models. Survival probabilities are calculated using life tables.

Patient Distribution

- 20 sites in 5 countries with randomization of 110 patients completed in August 2013 and distributed: Spain 45%; UK 29%; Italy 19%; Cyprus 5% and Ireland 2%. The treatment phase of the main study ended in June 2014.
- 12 patients were enrolled in the follow-up sub study at 8 sites, with patient distribution: Spain 6; UK 4; Italy 2.

Analysis and Reporting of Results

- Efficacy endpoint of primary interest was overall survival (OS).
- OS reported for Intent-to-treat (ITT), per protocol (PP) sets and metastatic sub-groups of these. PFS reported for ITT and metastatic sub-group.
- Safety and tolerability endpoints analyzed using the Safety Set.
- Survival data for the sub study with a cut-off date of April 25, 2015.

RESULTS

Figure 1: Overall Survival Kaplan–Meier Curves for ITT population

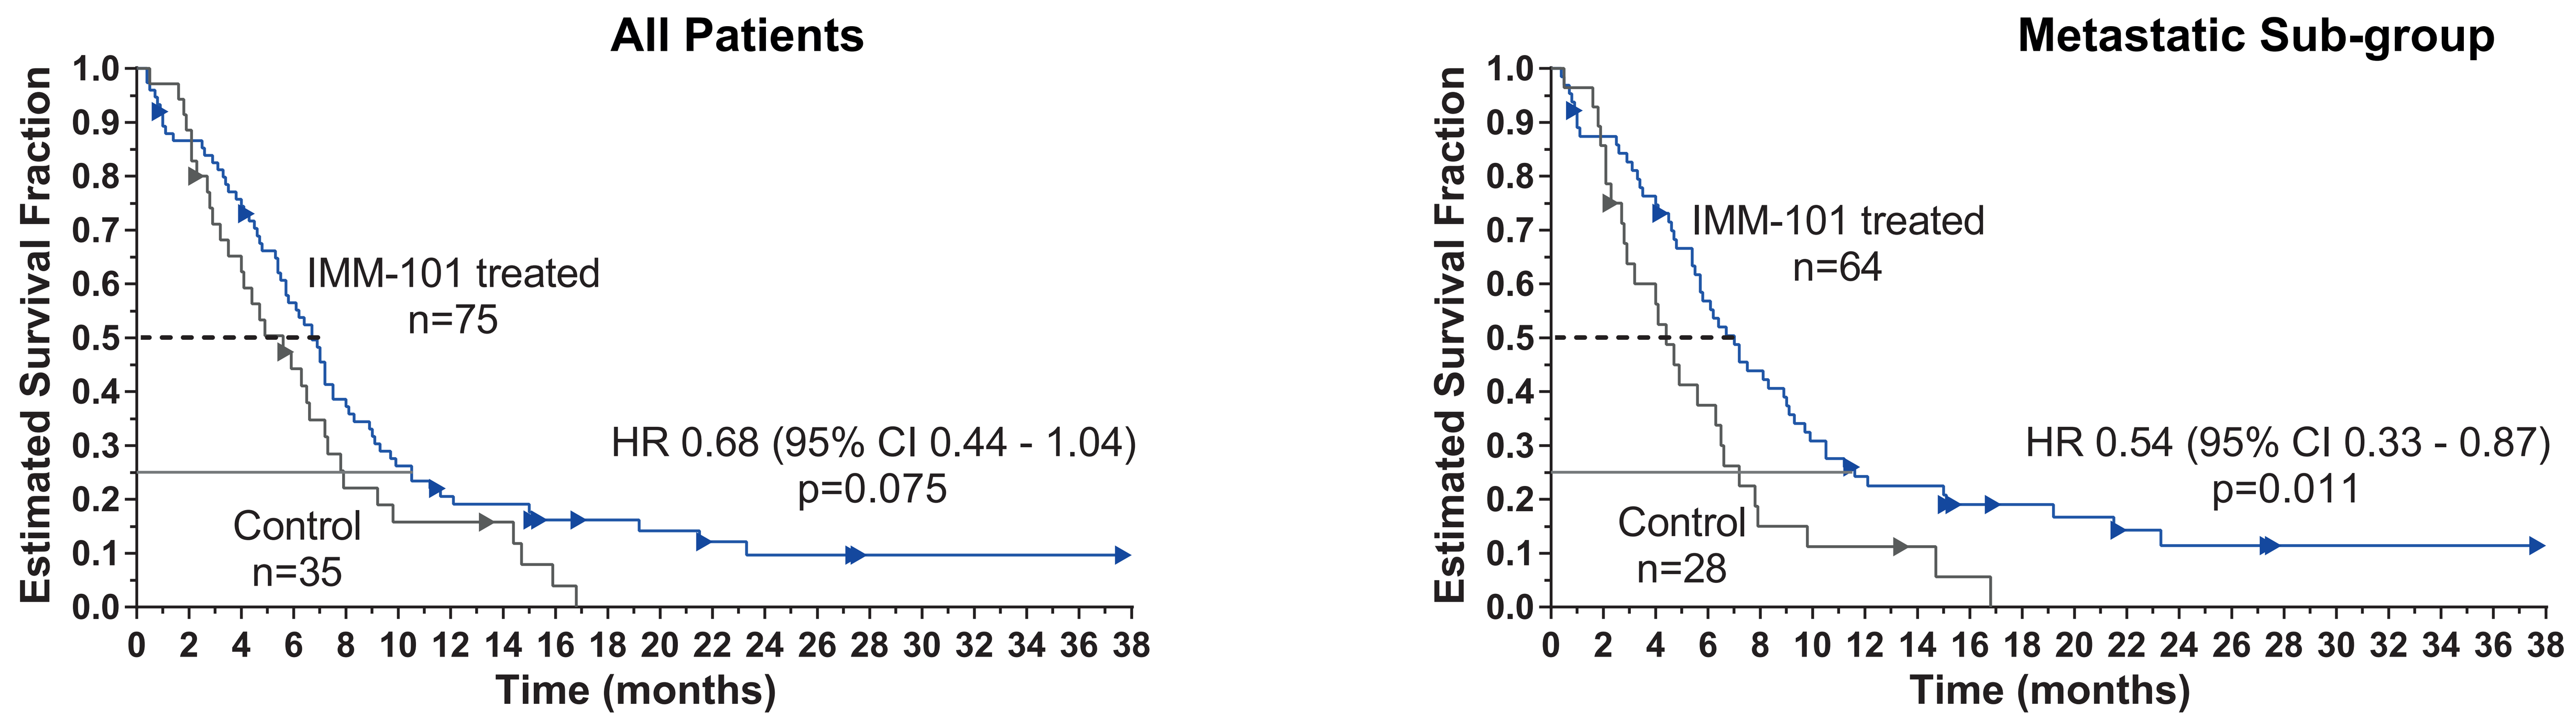
All Patients
Estimated Survival Fraction
IMM-101 treated
n=75
HR 0.68 (95% CI 0.44 - 1.04)
p=0.075
Control
n=35
Time (months)
Metastatic Sub-group
IMM-101 treated
n=64
HR 0.54 (95% CI 0.33 - 0.87)
p=0.011
Control
n=28

Figure 2: Overall Survival Kaplan–Meier Curves for PP population

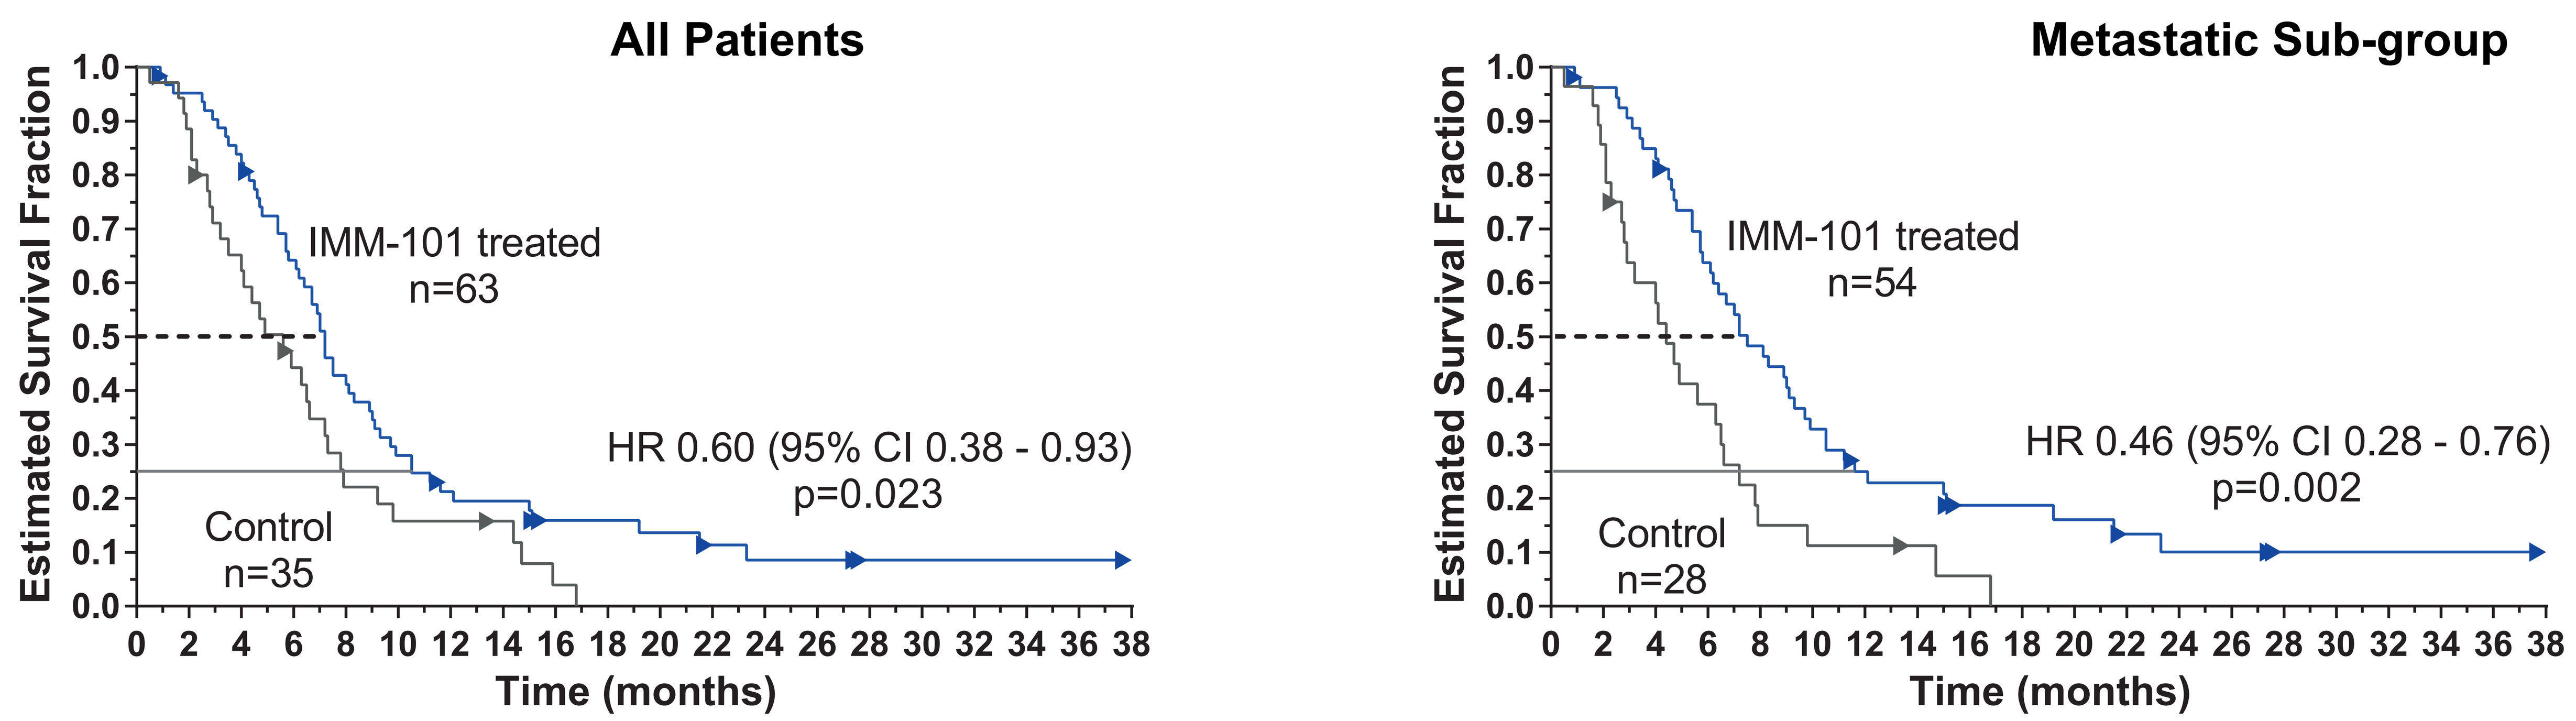
All Patients
Estimated Survival Fraction
IMM-101 treated
n=63
HR 0.60 (95% CI 0.38 - 0.93)
p=0.023
Control
n=35
Time (months)
Metastatic Sub-group
IMM-101 treated
n=54
HR 0.46 (95% CI 0.28 - 0.76)
p=0.002
Control
n=28

Figure 3: Survival Probability at 12, 18 and 24 months for ITT population

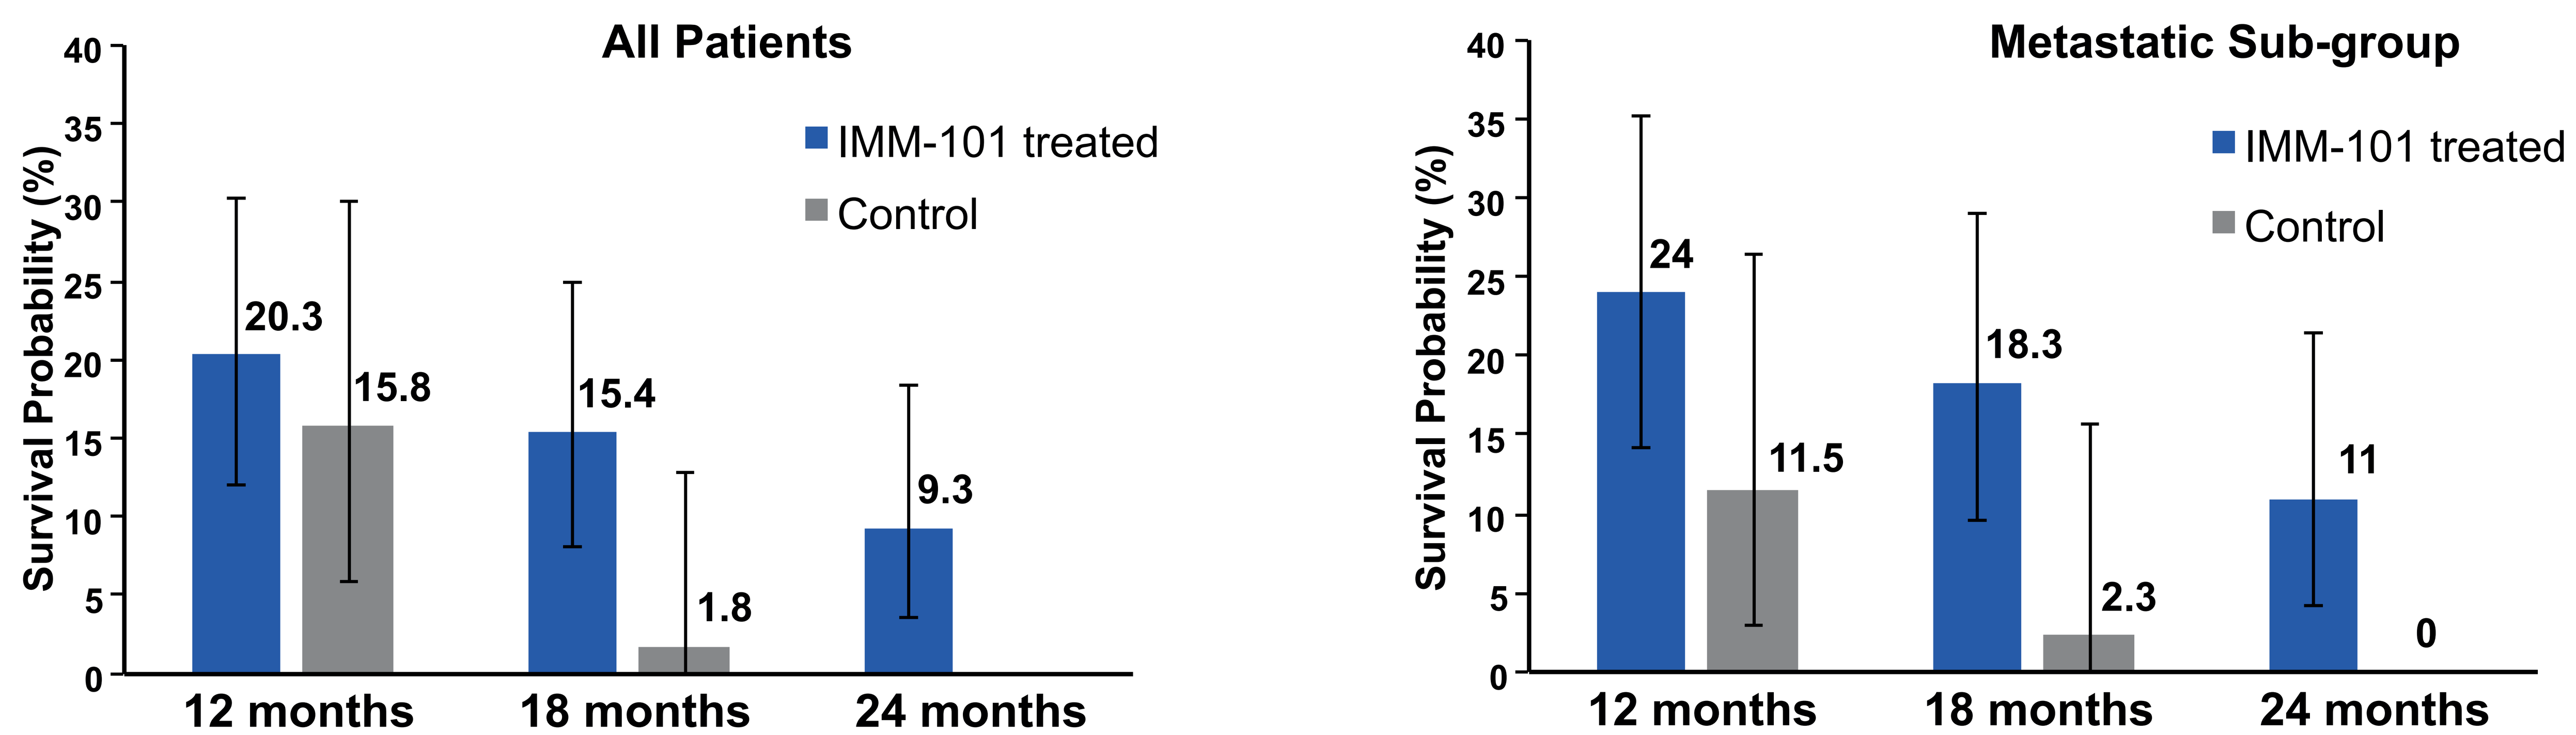
All Patients
Survival Probability (%)
IMM-101 treated
Control
20.3
15.8
15.4
1.8
9.3
12 months
18 months
24 months
Metastatic Sub-group
24
11.5
18.3
2.3
11
0

CONCLUSIONS

- Clinically meaningful increases in OS were demonstrated in IMM-101 treated patients with metastatic disease.
- The overall shape of the IMM-101 Kaplan-Meier curves is characteristic of immunotherapy agents (McDermott *et al.*, 2014).
- The continued separation of the Kaplan-Meier curves to 24 months and the survival probabilities at 12, 18 and 24 months are indicative of a durable response in some patients.
- The incidence of grade 3 or higher adverse events, being similar for both treatment groups, indicates there is no additional burden of adverse events in IMM-101 treated patients above that relating to chemotherapy or the underlying disease. Characteristic immune-related toxicities were not observed.
- The separation in the tail of the Kaplan-Meier curves previously reported at 12 months is amplified at 18 and 24 months. The IMM-101 treated curve then reaches a plateau, suggesting there are more durable responses in a small proportion of patients, consistent with observations of the effects of immunotherapy for other cancers, but not predicted in advanced pancreatic cancer.

Survival extensions, durable responses and a favourable safety profile mean this proof of concept study has exceeded its objectives.

This is a simplified version of a poster presented in May 2015 at the ASCO Annual Meeting, Chicago, USA

REFERENCES

Elia A *et al.*, 2013, J Immunother Cancer 1: Sup 1, P215

Stebbing J *et al.*, 2012. Ann Oncol 23:1314

Dalgleish A G *et al.*, ASCO meeting library, Gastrointestinal Cancers Symposium, 2015

McDermott *et al.*, Cancer Treatment Reviews 40 (2014) 1056-1064